Tivozanib hydrochloride pharmacokinetic/pharmacodynamic analysis of blood pressure and soluble vascular endothelial growth factor receptor 2 (sVEGFR2) in patients with advanced renal cell carcinoma

Dmitry A. Nosov,[1] Robert J. Motzer,[2] John Loewy,[3] Lee Hodge,[3] Brooke Esteves,[4] Anna Berkenblit,[4] Wei Yin,[4] Kevin Dykstra,[3] Thomas E. Hutson,[5] Monette M. Cotreau[4]

[1]N.N. Blokhin Cancer Research Center, under the Russian Academy of Medical Sciences, Clinical Pharmacology & Chemotherapy, Moscow, Russia; [2]Memorial Sloan-Kettering Cancer Center, New York, NY, USA; [3]qPharmetra, Andover, MA, USA; [4]AVEO Oncology, Cambridge, MA, USA; [5]Texas Oncology–Baylor Charles A. Sammons Cancer Center, Dallas, TX, USA

Introduction

- Tivozanib hydrochloride (tivozanib) is a novel, potent, selective, long half-life tyrosine kinase inhibitor of vascular endothelial growth factor receptors (VEGFRs) 1, 2, and 3, demonstrating activity in advanced renal cell carcinoma (RCC) in Phase II and III trials[1–3]
 - In healthy volunteers, exposure (maximum serum concentration and area under the curve) to tivozanib generally increases in a dose-proportional manner, and accumulation at steady state is approximately 6–7 times single-dose levels[4]
 - Tivozanib also has a long half-life of 4.5–5.1 days[4,5]
- The relationship between tivozanib exposure, blood pressure, and sVEGFR2 was explored. Hypertension and sVEGFR are known as potentially relevant predictive biomarkers of activity VEGFR-inhibitors and clinical outcome[6–8]

Methods

- Pharmacokinetic, blood pressure, and sVEGFR2 data from tivozanib-treated RCC patients from a Phase II (n=21) and a Phase III (n=259) study were pooled and analyzed
- The Phase II study (AV-951-07-201; NCT00502307) was a randomized, placebo-controlled discontinuation trial to determine the safety and efficacy of tivozanib, and the Phase III study (AV-951-09-301; NCT01030783) was a randomized controlled, multicenter, open-label study to compare tivozanib with sorafenib
- In both studies, patients were treated with 1.5 mg tivozanib daily for 21 days followed by a 7-day rest (28-day treatment cycle). Patients in both trials were treated for multiple cycles
- The following clinical and biological endpoints were analyzed based on a time-dependent relationship:
 - sVEGFR2
 - Diastolic blood pressure
 - Systolic blood pressure
- Blood pressure measurements taken on Cycle 1 Day 1 (predose baseline) and on Cycle 1 Day 15, Cycle 2 Day 1, and Cycle 3 Day 1 were used in this analysis. Measurements were rounded to the nearest 5 mm Hg, and the analysis focused on blood pressure shifts in 5 mm Hg increments
- Serum samples for sVEGFR2 (Phase III study only) were collected on Cycle 1 Day 1 (predose) and on Cycle 1 Day 15, Cycle 2 Day 1, and Cycle 2 Days 22–28
- Models of drug exposure as predictors of longitudinal changes in sVEGFR2 were constructed by non-linear, mixed-effects modeling
 - sVEGFR2 and blood pressure were considered continuous endpoints and were binned into average concentration (C_{avg}) quartiles containing an equal number of patients in four exposure categories to examine the interrelationships
 - Analysis was carried out using R (V2.14.0, R Development Core Team [2008]), or NONMEM (Version 7.2, ICON Development Solutions, Ellicott City, MD, USA)

Results

Tivozanib Exposure and Blood Pressure

- Among tivozanib-treated patients, diastolic blood pressure increased a median of 5 mm Hg relative to Day 1 at all post-dose time points in the first two treatment cycles. Diastolic blood pressure vs time over the first two treatment cycles in each study for tivozanib patients is shown in **Figure 1**
- Systolic blood pressure did not show a significant change from baseline after the start of treatment, and no treatment-related or other covariate effects on systolic blood pressure could be identified

Figure 1. Diastolic blood pressure vs time for the first two treatment cycles for tivozanib-treated patients.

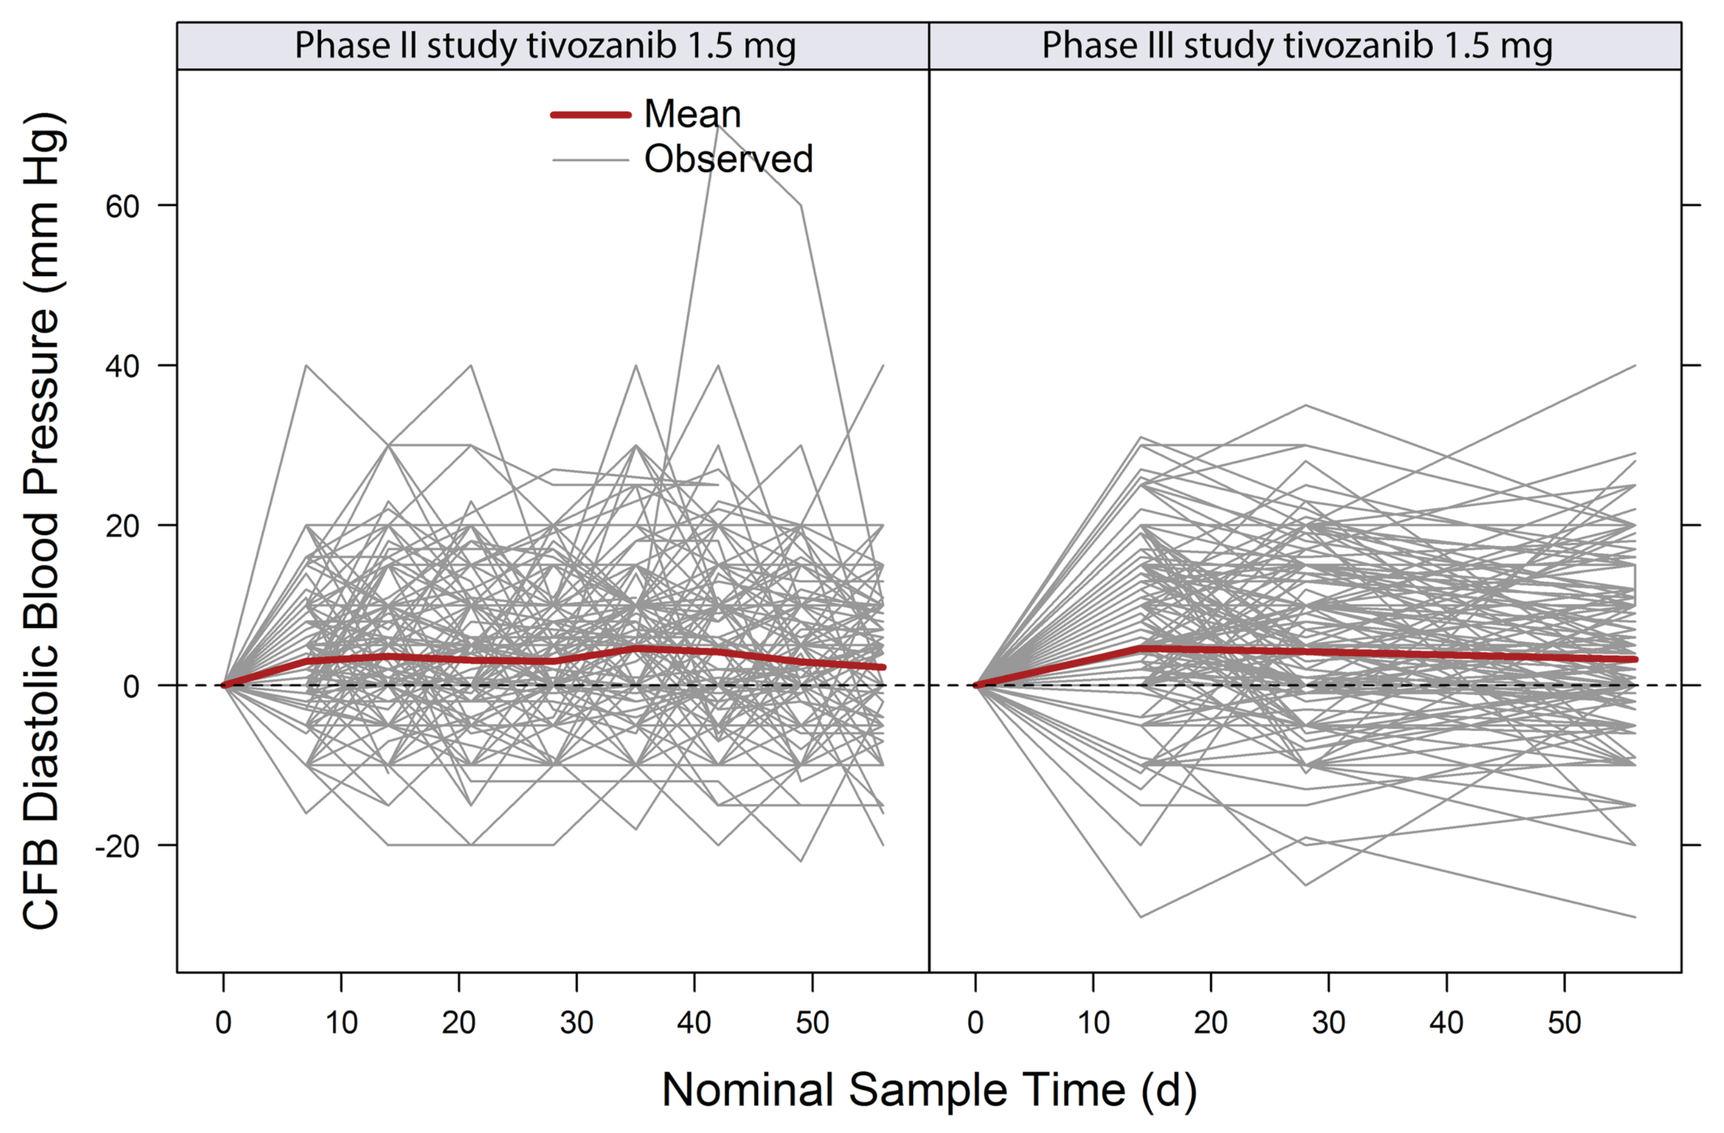
Phase II study tivozanib 1.5 mg
Phase III study tivozanib 1.5 mg
Mean
Observed
CFB Diastolic Blood Pressure (mm Hg)
Nominal Sample Time (d)

CFB, change from baseline.

- No significant relationship was seen between the measured changes in diastolic blood pressure and the differences in tivozanib exposure
- The change in diastolic blood pressure as a function by study day after the start of tivozanib treatment is shown in **Table 1** and **Figure 2**

Table 1. Change in Diastolic Blood Pressure (mm Hg) by Cycle and Study Day

Cycle and Study Day	N	Min	25th percentile	Median	Mean	75th percentile	Max	Standard Deviation
Cycle 1 Day 15	278	–30.00	0.00	5.00	4.66	10.00	30.00	9.37
Cycle 2 Day 1	278	–25.00	0.00	5.00	4.30	10.00	35.00	9.40
Cycle 3 Day 1	255	–30.00	0.00	0.00	3.35	10.00	40.00	9.74

Figure 2. Effect of tivozanib exposure on change in diastolic blood pressure at Cycle 1 Day 15.

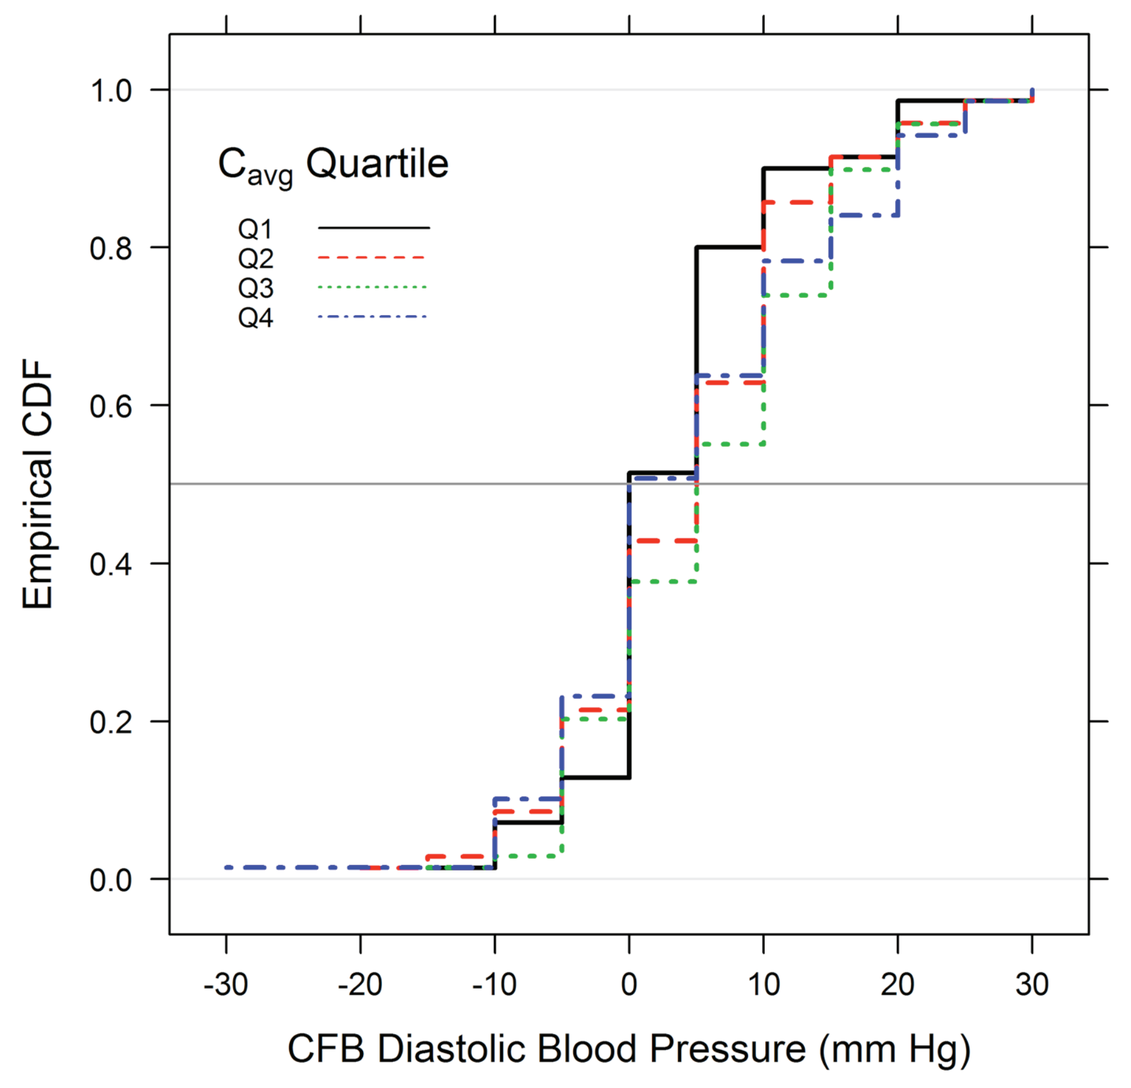
C_{avg} Quartile
Q1
Q2
Q3
Q4
Empirical CDF
CFB Diastolic Blood Pressure (mm Hg)

CDF, cumulative distribution function; CFB, change from baseline.

- Similarly there was no significant relationship between the measured changes in systolic blood pressure and the differences in tivozanib exposure. Changes in systolic blood pressure as a function by study day after the start of tivozanib treatment is shown in **Table 2** and **Figure 3**

Table 2. Change in Systolic Blood Pressure (mm Hg) by Study Day

Cycle and Study Day	N	Min	25th percentile	Median	Mean	75th percentile	Max	Standard Deviation
Cycle 1 Day 15	278	–30	–5.00	0.00	4.41	10.00	65.00	11.84
Cycle 2 Day 1	278	–35	–5.00	0.00	3.60	10.00	40.00	12.33
Cycle 3 Day 1	255	–50	–5.00	0.00	2.25	10.00	80.00	14.73

Figure 3. Effect of tivozanib exposure on change in systolic blood pressure at Cycle 1 Day 15.

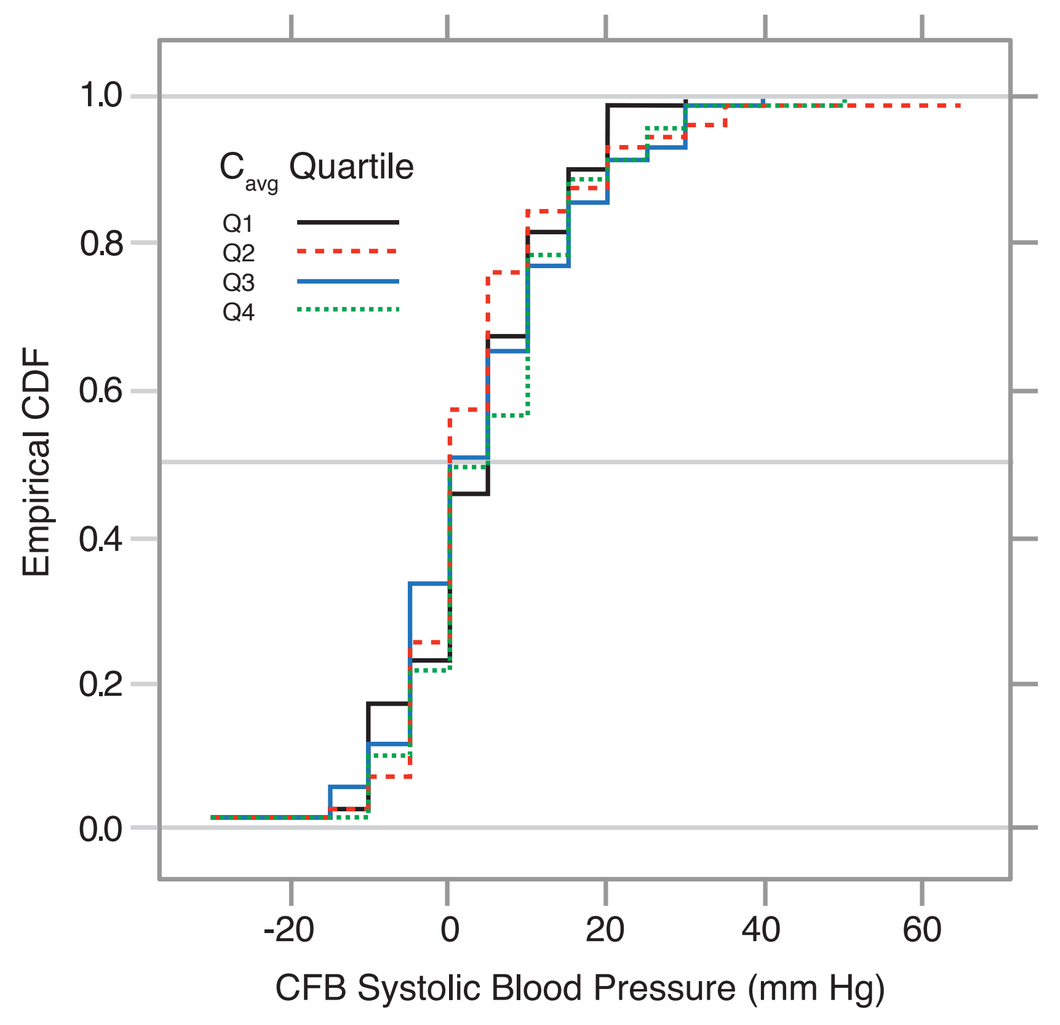
C_{avg} Quartile
Q1
Q2
Q3
Q4
Empirical CDF
CFB Systolic Blood Pressure (mm Hg)

CDF, cumulative distribution function; CFB, change from baseline.

- No temporal exposure-response relationship could be identified among those patients treated with tivozanib, and no other covariate relationships were identified for diastolic or systolic blood pressure

Pharmacokinetics and sVEGFR2

- sVEGFR2 declined as a non-linear function of time from Day 1 to Cycle 2 Days 22–28 among tivozanib-treated patients, and this decrease appeared saturable with time. The curvilinear decrease in sVEGFR2 over time is shown in **Figure 4**

Figure 4. Mean change in sVEGFR2 vs time in tivozanib-treated patients.

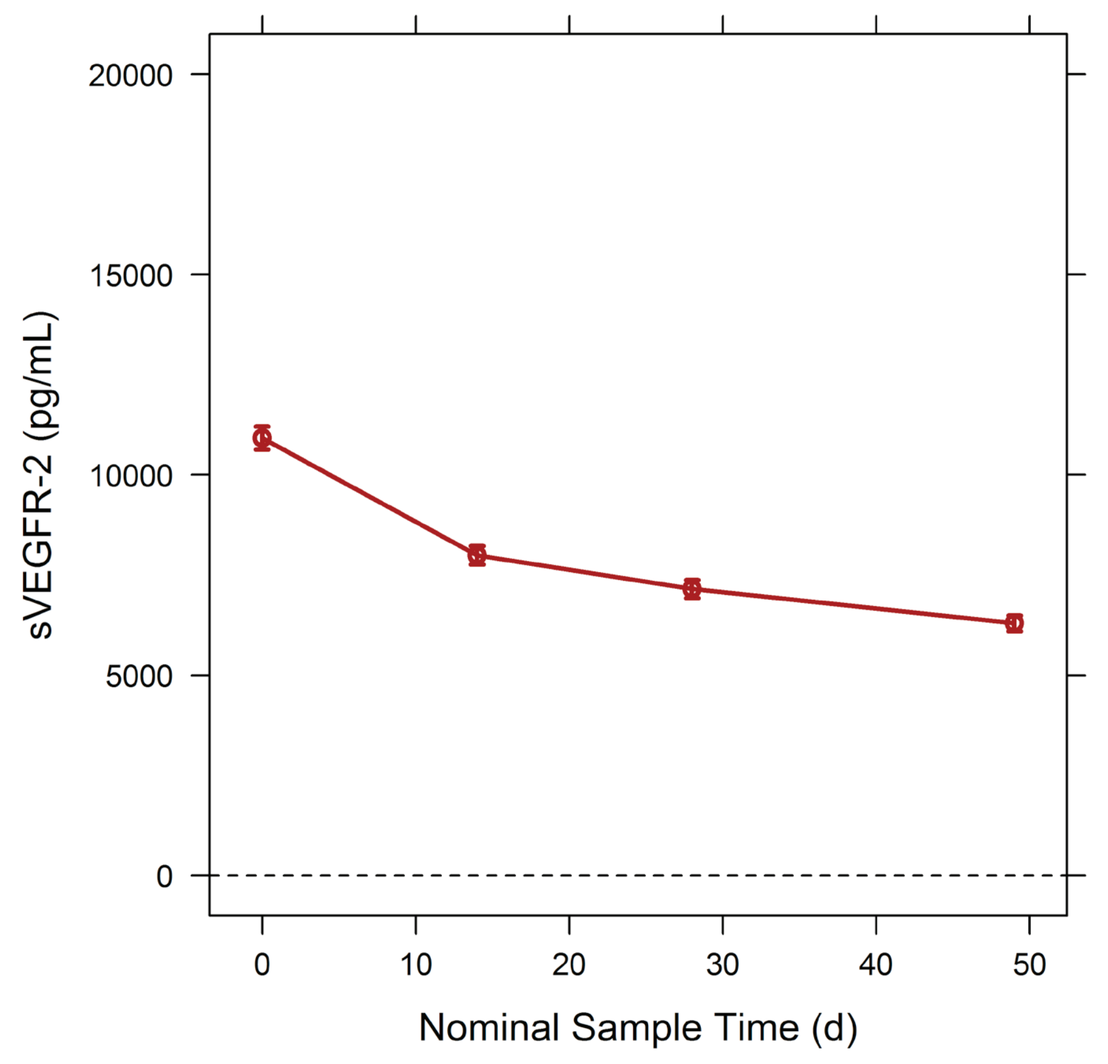
sVEGFR-2 (pg/mL)
Nominal Sample Time (d)

- The decrease in sVEGFR2 was greater as C_{avg} increased. The size of decrease in sVEGFR2 drug effect increased approximately 6% for each 10 ng/mL increase in C_{avg} (**Figure 5**)

Figure 5. Change in sVEGFR2 as a function of C_{avg}.

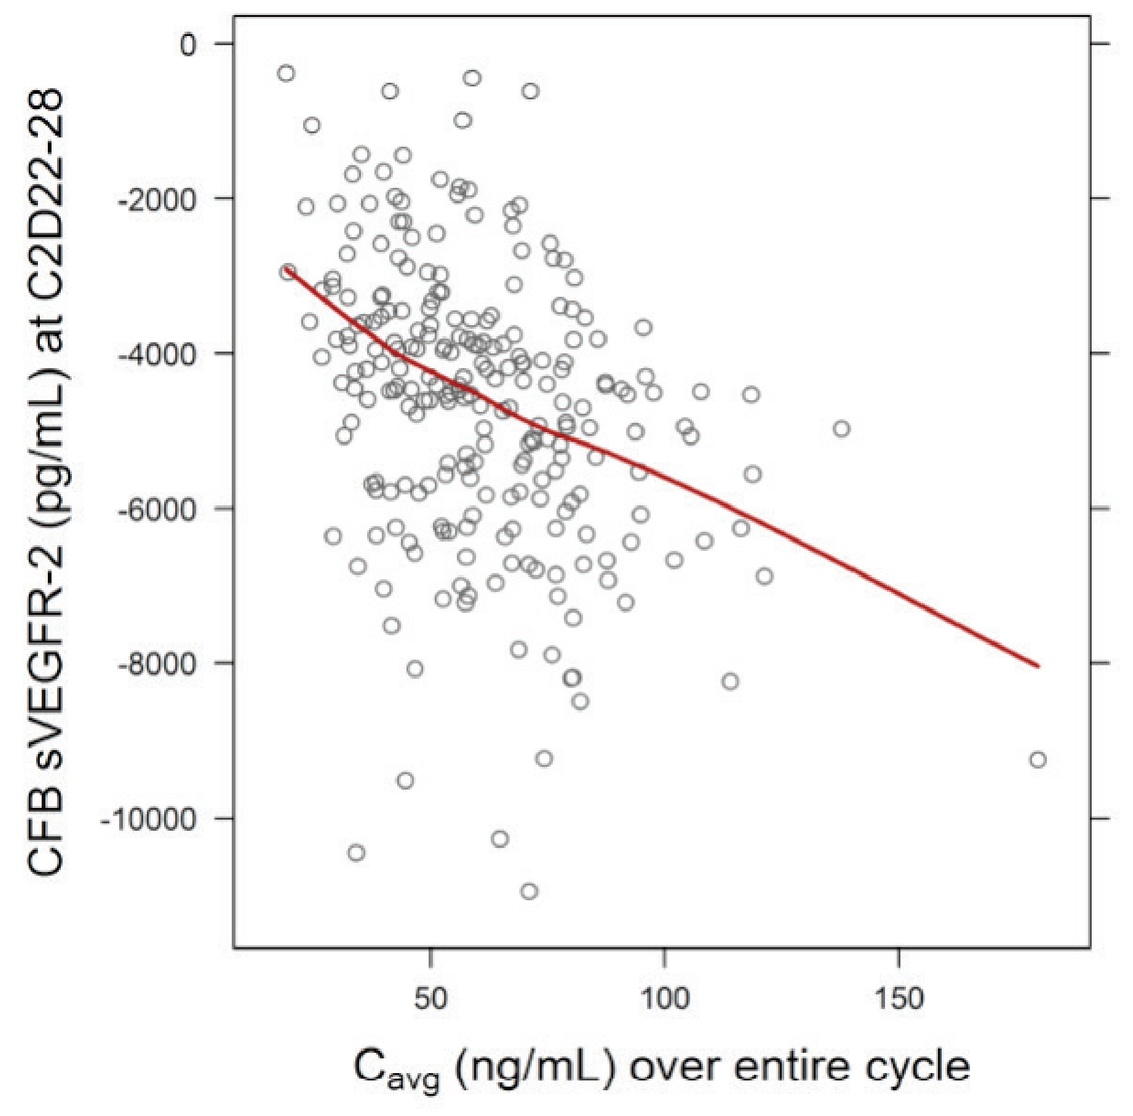
CFB sVEGFR-2 (pg/mL) at C2D22-28
C_{avg} (ng/mL) over entire cycle

CFB, change from baseline.

Conclusions

- Pharmacokinetic/Pharmacodynamic analysis of data from patients treated with tivozanib in Phase II and III studies showed that patients had a median increase in diastolic blood pressure of 5 mm Hg on Cycle 1 Day 15 and Cycle 2 Day 1 compared with baseline
- Levels of serum sVEGFR2 were found to decrease with time, and the effect size increased with tivozanib exposure. These findings are consistent with previous reports that decreases of sVEGFR may serve as a pharmacodynamic marker of VEGFR inhibition
- A significant association of tivozanib exposure and blood pressure is likely, but has not been found in the present analysis. This might be due to infrequent monitoring of blood pressure in the Phase III study

References

1. Nakamura K, Taguchi E, Miura T *et al.* KRN951, a highly potent inhibitor of vascular endothelial growth factor receptor tyrosine kinases, has antitumor activities and affects functional vascular properties. *Cancer Res* 2006;66:9134–9142.
2. Nosov DA, Esteves B, Lipatov ON *et al.* Antitumor activity and safety of tivozanib (AV-951) in a Phase II randomized discontinuation trial in patients with renal cell carcinoma. *J Clin Oncol* 2012;30:1678–1685.
3. Motzer RJ, Eisen T, Bondarenko IN *et al.* Tivozanib versus sorafenib as initial targeted therapy for patients with advanced renal cell carcinoma: Results from a phase III randomized, open-label, multicenter trial. *J Clin Oncol* 2012;30(suppl):Abstract 4501.
4. Aveo Oncology. Data on file. 2012.
5. Cotreau MM, King TA, Massmanian L *et al.* The effect of food on the pharmacokinetics of tivozanib. In: Proceedings of the 103rd Annual Meeting of the American Association for Cancer Research; 2012 Mar 31–Apr 4; Chicago, IL. Philadelphia (PA): AACR; *Cancer Res* 2012;72(8 suppl):Abstract nr 752.
6. De Primo SE, Bello CL, Smeraglia J *et al.* Circulating protein biomarkers of pharmacodynamic activity of sunitinib in patients with metastatic renal cell carcinoma: modulation of VEGF and VEGF related proteins. *J Transl Med* 2007;5:32.
7. Hutson T, Davis ID, Macheils JH *et al.* Biomarker analysis and final efficacy and safety results of a phase II renal cell carcinoma trial with pazopanib (GW786034), a multi-kinase angiogenesis inhibitor. *J Clin Oncol* 2008;26:Abstract 5046.
8. Rini BI, Michaelson D, Rosenberg JE et al. Antitumor activity and biomarker analysis of sunitinib in patients with bevacizumab-refracory metastatic renal cell carcinoma. *J Clin Oncol* 2008;26:3743–3748.

Acknowledgments

This study was supported by AVEO Oncology and Astellas. AVEO Oncology and Astellas are parties to a collaboration agreement for the co-development of tivozanib. Editorial assistance was provided by Isabelle Leach, MBChB, Chameleon Communications International, and was funded by AVEO Oncology and Astellas.

POSTER PRESENTED AT THE 2012 EUROPEAN SOCIETY FOR MEDICAL ONCOLOGY (ESMO) ANNUAL MEETING; VIENNA, AUSTRIA; SEPTEMBER 28–OCTOBER 2, 2012.

Corresponding author: Dmitry A. Nosov (nosov@mail.ru)