5062

TIVO-3: Final OS analysis of a randomized, controlled, multi-center, open-label phase 3 study to compare tivozanib to sorafenib in subjects with metastatic renal cell carcinoma (RCC)

Sumanta K. Pal,[1] Bernard Escudier,[2] Michael B. Atkins,[3] Thomas E. Hutson,[4] Camillo Porta,[5] Elena Verzoni,[6] Michael N. Needle,[7] David F. McDermott,[8] Brian I. Rini[9]

[1]Kidney Cancer Program, City of Hope National Medical Center, Duarte, CA, USA; [2]Department of Oncology Medicine, Gustave Roussy Cancer Campus, Villejuif, France; [3]Department of Medical Oncology, Georgetown Lombardi University Hospital, Lombardi Comprehensive Cancer Center, Washington, DC, USA; [4]Urologic Oncology, Baylor Sammons Cancer Center-Texas Oncology, Dallas, TX, USA; [5]Department of Internal Medicine, University of Pavia, Division of Translational Oncology, IRCCS Istituti Clinici Scientifici Maugeri, Pavia, Italy; [6]Department of Medical Oncology, Fondazione IRCCS Istituto Nazionale Tumori, Milan, Italy; [7]AVEO Oncology; Boston, MA, USA; [8]Beth Israel Deaconess Medical Center, Boston, MA, USA; [9]Vanderbilt University Medical Center; Nashville, TN, USA

Background

- The treatment of patients with renal cell carcinoma (RCC) has advanced with the advent of anti-angiogenic drugs targeting the vascular endothelial growth factor receptor (VEGFR)[1-4]
- Tivozanib is a potent, highly selective VEGFR TKI with a long half-life and an ability to downregulate T-regulatory cells
- Tivozanib is approved by the European Medicines Agency for the first-line treatment of adult patients with RCC[5]
 - Tivozanib was developed to optimize the VEGFR blockade while minimizing off-target toxicities[6,7]
- The TIVO-3 study was designed to assess the safety and efficacy of tivozanib versus sorafenib for third-line and fourth-line treatment of metastatic RCC[8]
 - In TIVO-3, a significant reduction in the risk of progression-free survival (PFS) was reported for tivozanib compared with sorafenib at a hazard ratio (HR) of 0.73 (95% confidence interval [CI]: 0.56, 0.94; *P*=0.016); median PFS was 5.6 months vs. 3.9 months[8]
 - Tivozanib treatment demonstrated a favorable safety profile
- Final OS results for the TIVO-3 study are presented herein, including an analysis of OS in prespecified patient subgroups

Methods

Study Design

- TIVO-3 (NCT02627963) is an open-label, randomized phase 3 study comparing tivozanib with sorafenib in patients with metastatic RCC (**Figure 1**)
- Patients were randomized 1:1 to receive tivozanib 1.5 mg once daily in 4-week cycles (21 days on treatment followed by 7 days off-treatment) or sorafenib 400 mg twice daily continuously

Figure 1. TIVO-3 Study Design

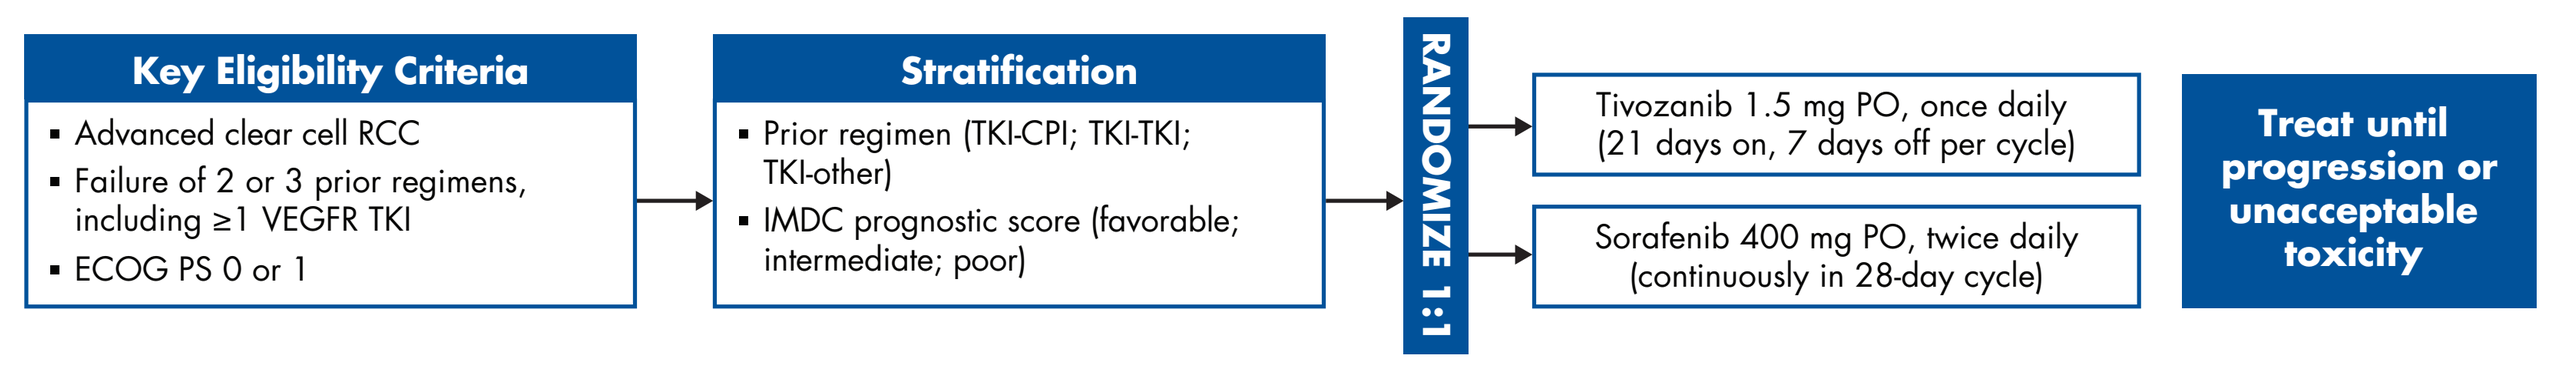

Key Eligibility Criteria
▪ Advanced clear cell RCC
▪ Failure of 2 or 3 prior regimens, including ≥1 VEGFR TKI
▪ ECOG PS 0 or 1
Stratification
▪ Prior regimen (TKI-CPI; TKI-TKI; TKI-other)
▪ IMDC prognostic score (favorable; intermediate; poor)
RANDOMIZE 1:1
Tivozanib 1.5 mg PO, once daily (21 days on, 7 days off per cycle)
Sorafenib 400 mg PO, twice daily (continuously in 28-day cycle)
Treat until progression or unacceptable toxicity

CPI=checkpoint inhibitor; ECOG PS=Eastern Cooperative Oncology Group performance status; IMDC=International Metastatic Renal Cell Carcinoma Database Consortium; PO=orally; RCC=renal cell carcinoma; TKI=tyrosine kinase inhibitor; VEGFR=vascular endothelial growth factor receptor.

Study Endpoints

- The primary endpoint was PFS, defined as the time from randomization to the first documentation of tumor progression by the independent review committee (radiologic progressive disease) according to the Response Evaluation Criteria in Solid Tumors version 1.1 (RECIST v1.1)
- Pre-specified secondary endpoints of the study included OS (defined as the time from randomization to the date of death due to any cause), objective response rate, duration of response, and safety
- In total, three prespecified assessments of OS were conducted (two interim analyses and one final analysis); data included herein are inclusive of the final OS analysis

Results

Patients

- Baseline demographics and disease characteristics were balanced among treatment groups and typical of those seen in patients with advanced RCC (**Table 1**)
 - Median time from initial diagnosis was 50 months for both treatment arms
 - Median duration of exposure was 197 days for the tivozanib group and 141 days for the sorafenib group

Table 1. Key Baseline Patient Demographics[8]

	Tivozanib (n=175)	Sorafenib (n=175)
Age, years	62 (34–88)	64 (30–90)
Sex		
Male	126 (72)	128 (73)
Female	49 (28)	47 (27)
IMDC risk category		
Favorable	34 (19)	36 (21)
Intermediate	109 (62)	105 (60)
Poor	32 (18)	34 (19)
Number of previous systemic therapies		
Two	108 (62)	104 (59)
Three	67 (38)	71 (41)
Previous therapies		
Two VEGFR TKIs	79 (45)	80 (46)
Checkpoint inhibitor (CPI) plus VEFGR TKI	47 (27)	44 (25)
VEGFR TKI plus other	49 (28)	51 (29)

Data are n (%) or median (range).
IMDC=International Metastatic Renal Cell Carcinoma Database Consortium; TKI=tyrosine kinase inhibitor; VEGFR=vascular endothelial growth factor receptor.

PFS

- As previously reported, at the October 2018 final analysis with a median follow-up of 19 months, 246 PFS events occurred
 - A significant reduction in the risk of PFS was observed for tivozanib compared with sorafenib with an HR of 0.73 (95% CI: 0.56, 0.94; *P*=0.016) and median PFS of 5.6 months vs. 3.9 months (**Figure 2**)[8]

Figure 2. PFS in the ITT Population[8]

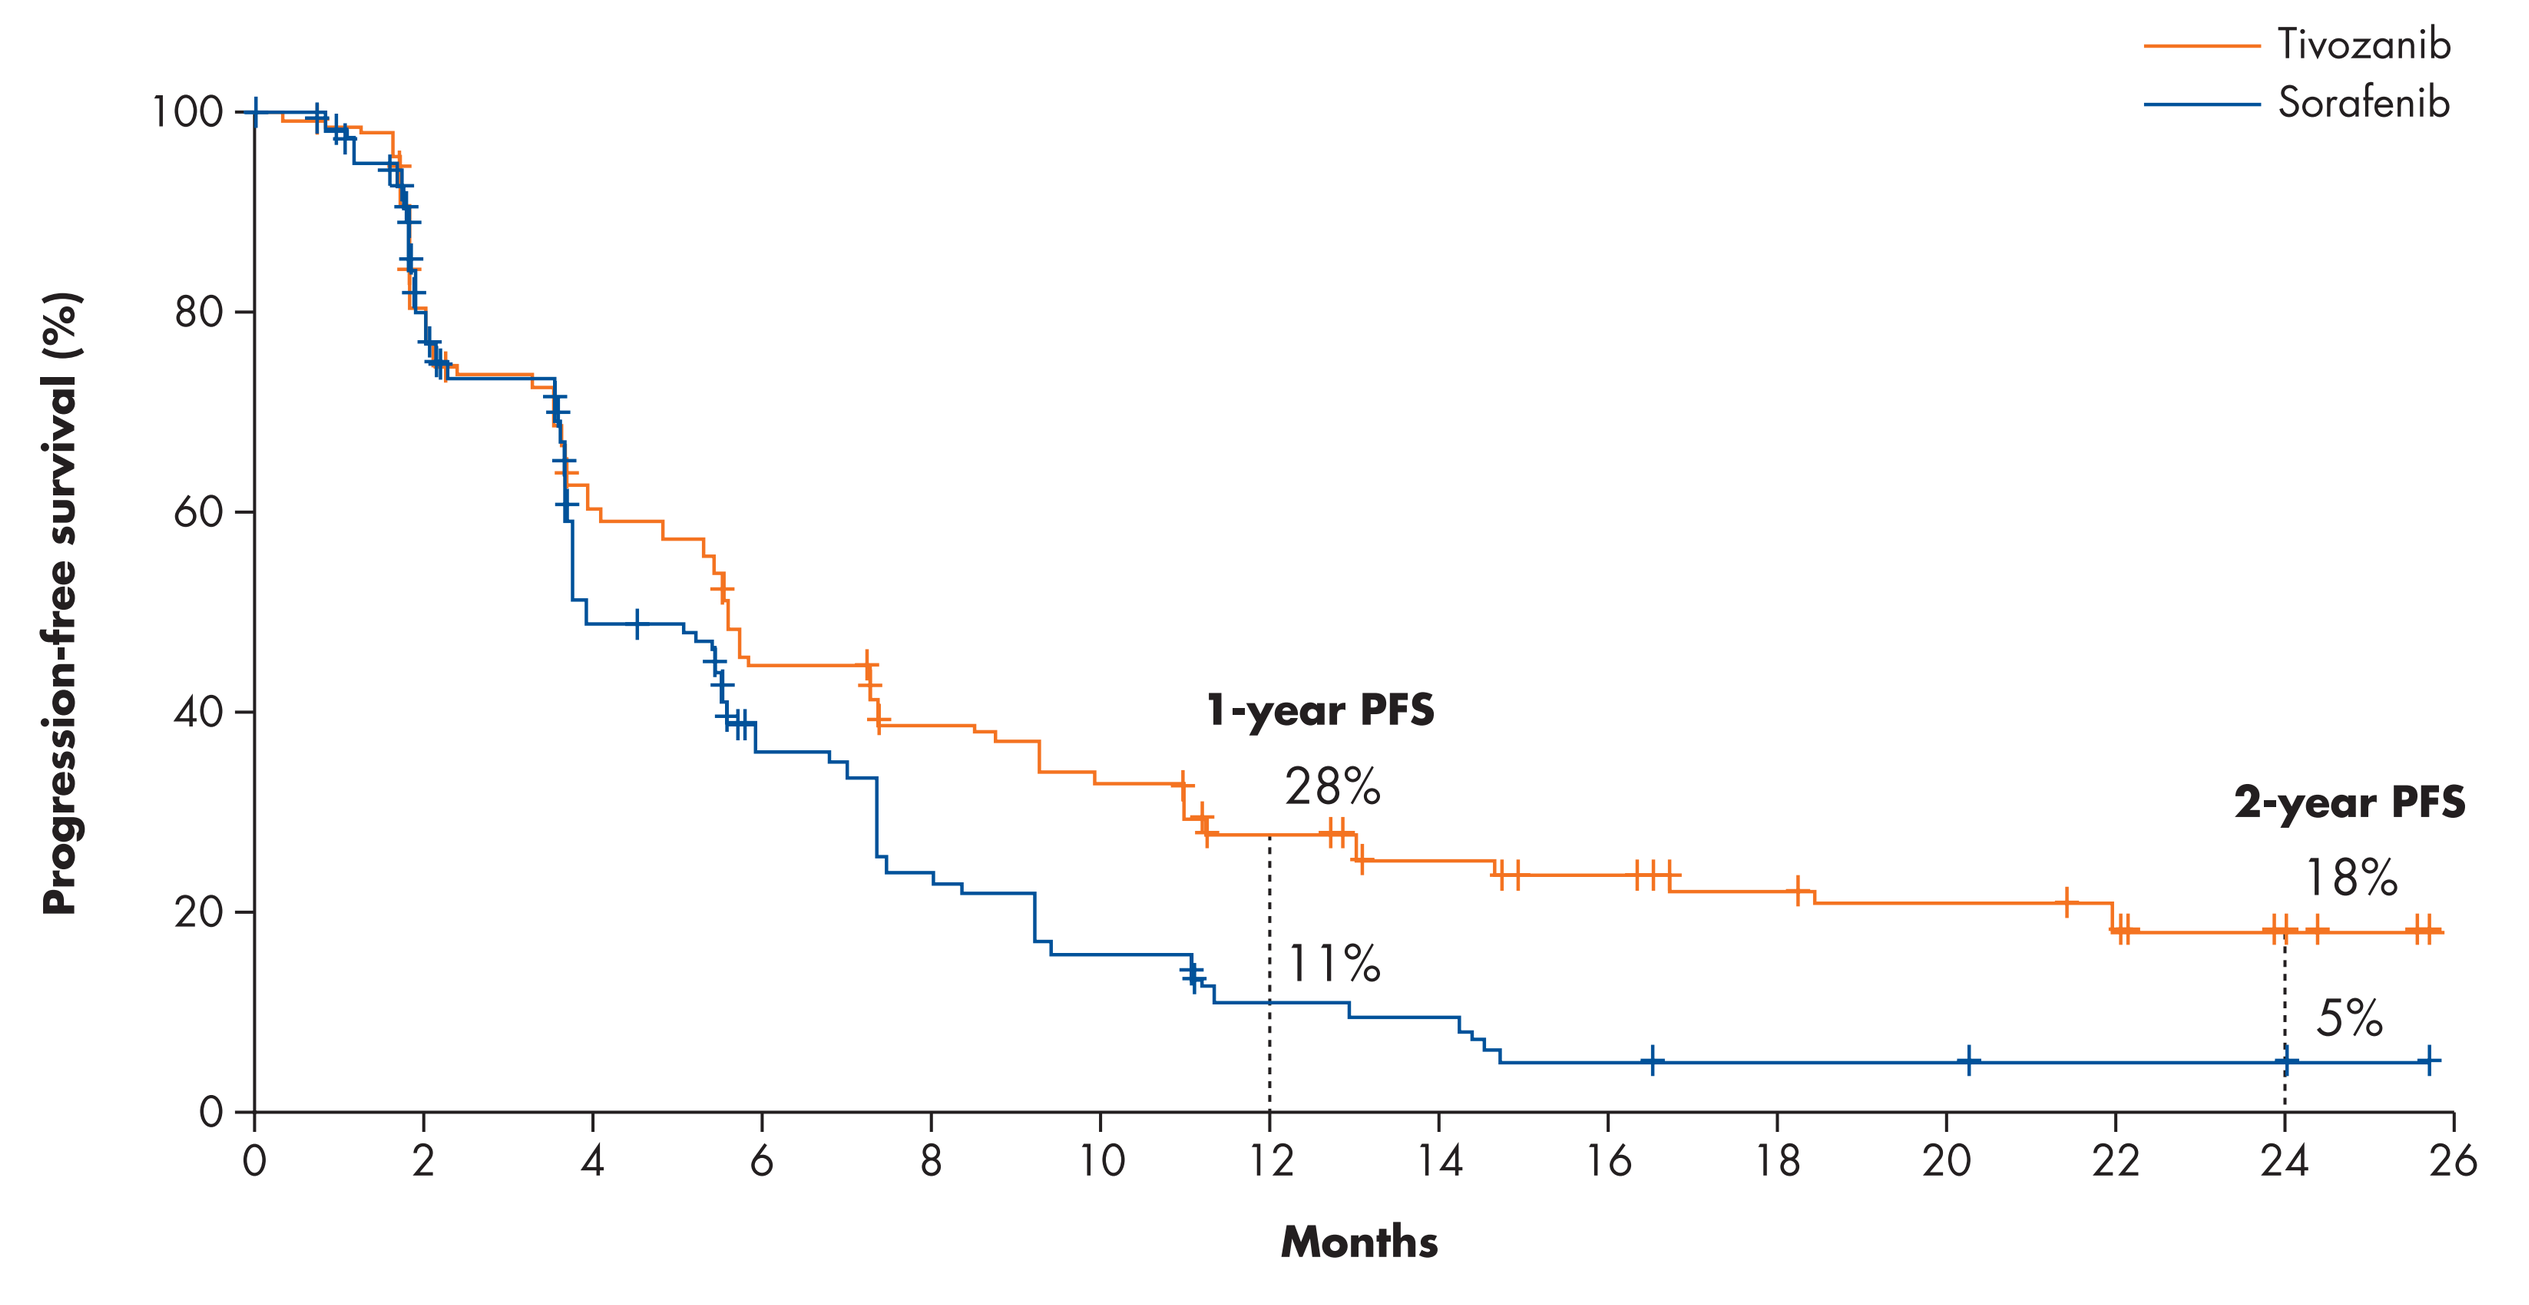

Tivozanib
Sorafenib
Progression-free survival (%)
100
80
60
40
20
0
1-year PFS
28%
11%
2-year PFS
18%
5%
0 2 4 6 8 10 12 14 16 18 20 22 24 26
Months

ITT=intent-to-treat.

- In patients who received a prior checkpoint inhibitor (CPI) plus VEGFR TKI or 2 prior VEGFR TKIs, a significant reduction in the risk of PFS was observed for tivozanib compared with sorafenib with an HR of 0.55 and 0.57, respectively (**Figure 3**)[8]

Figure 3. PFS by Patient Subgroups

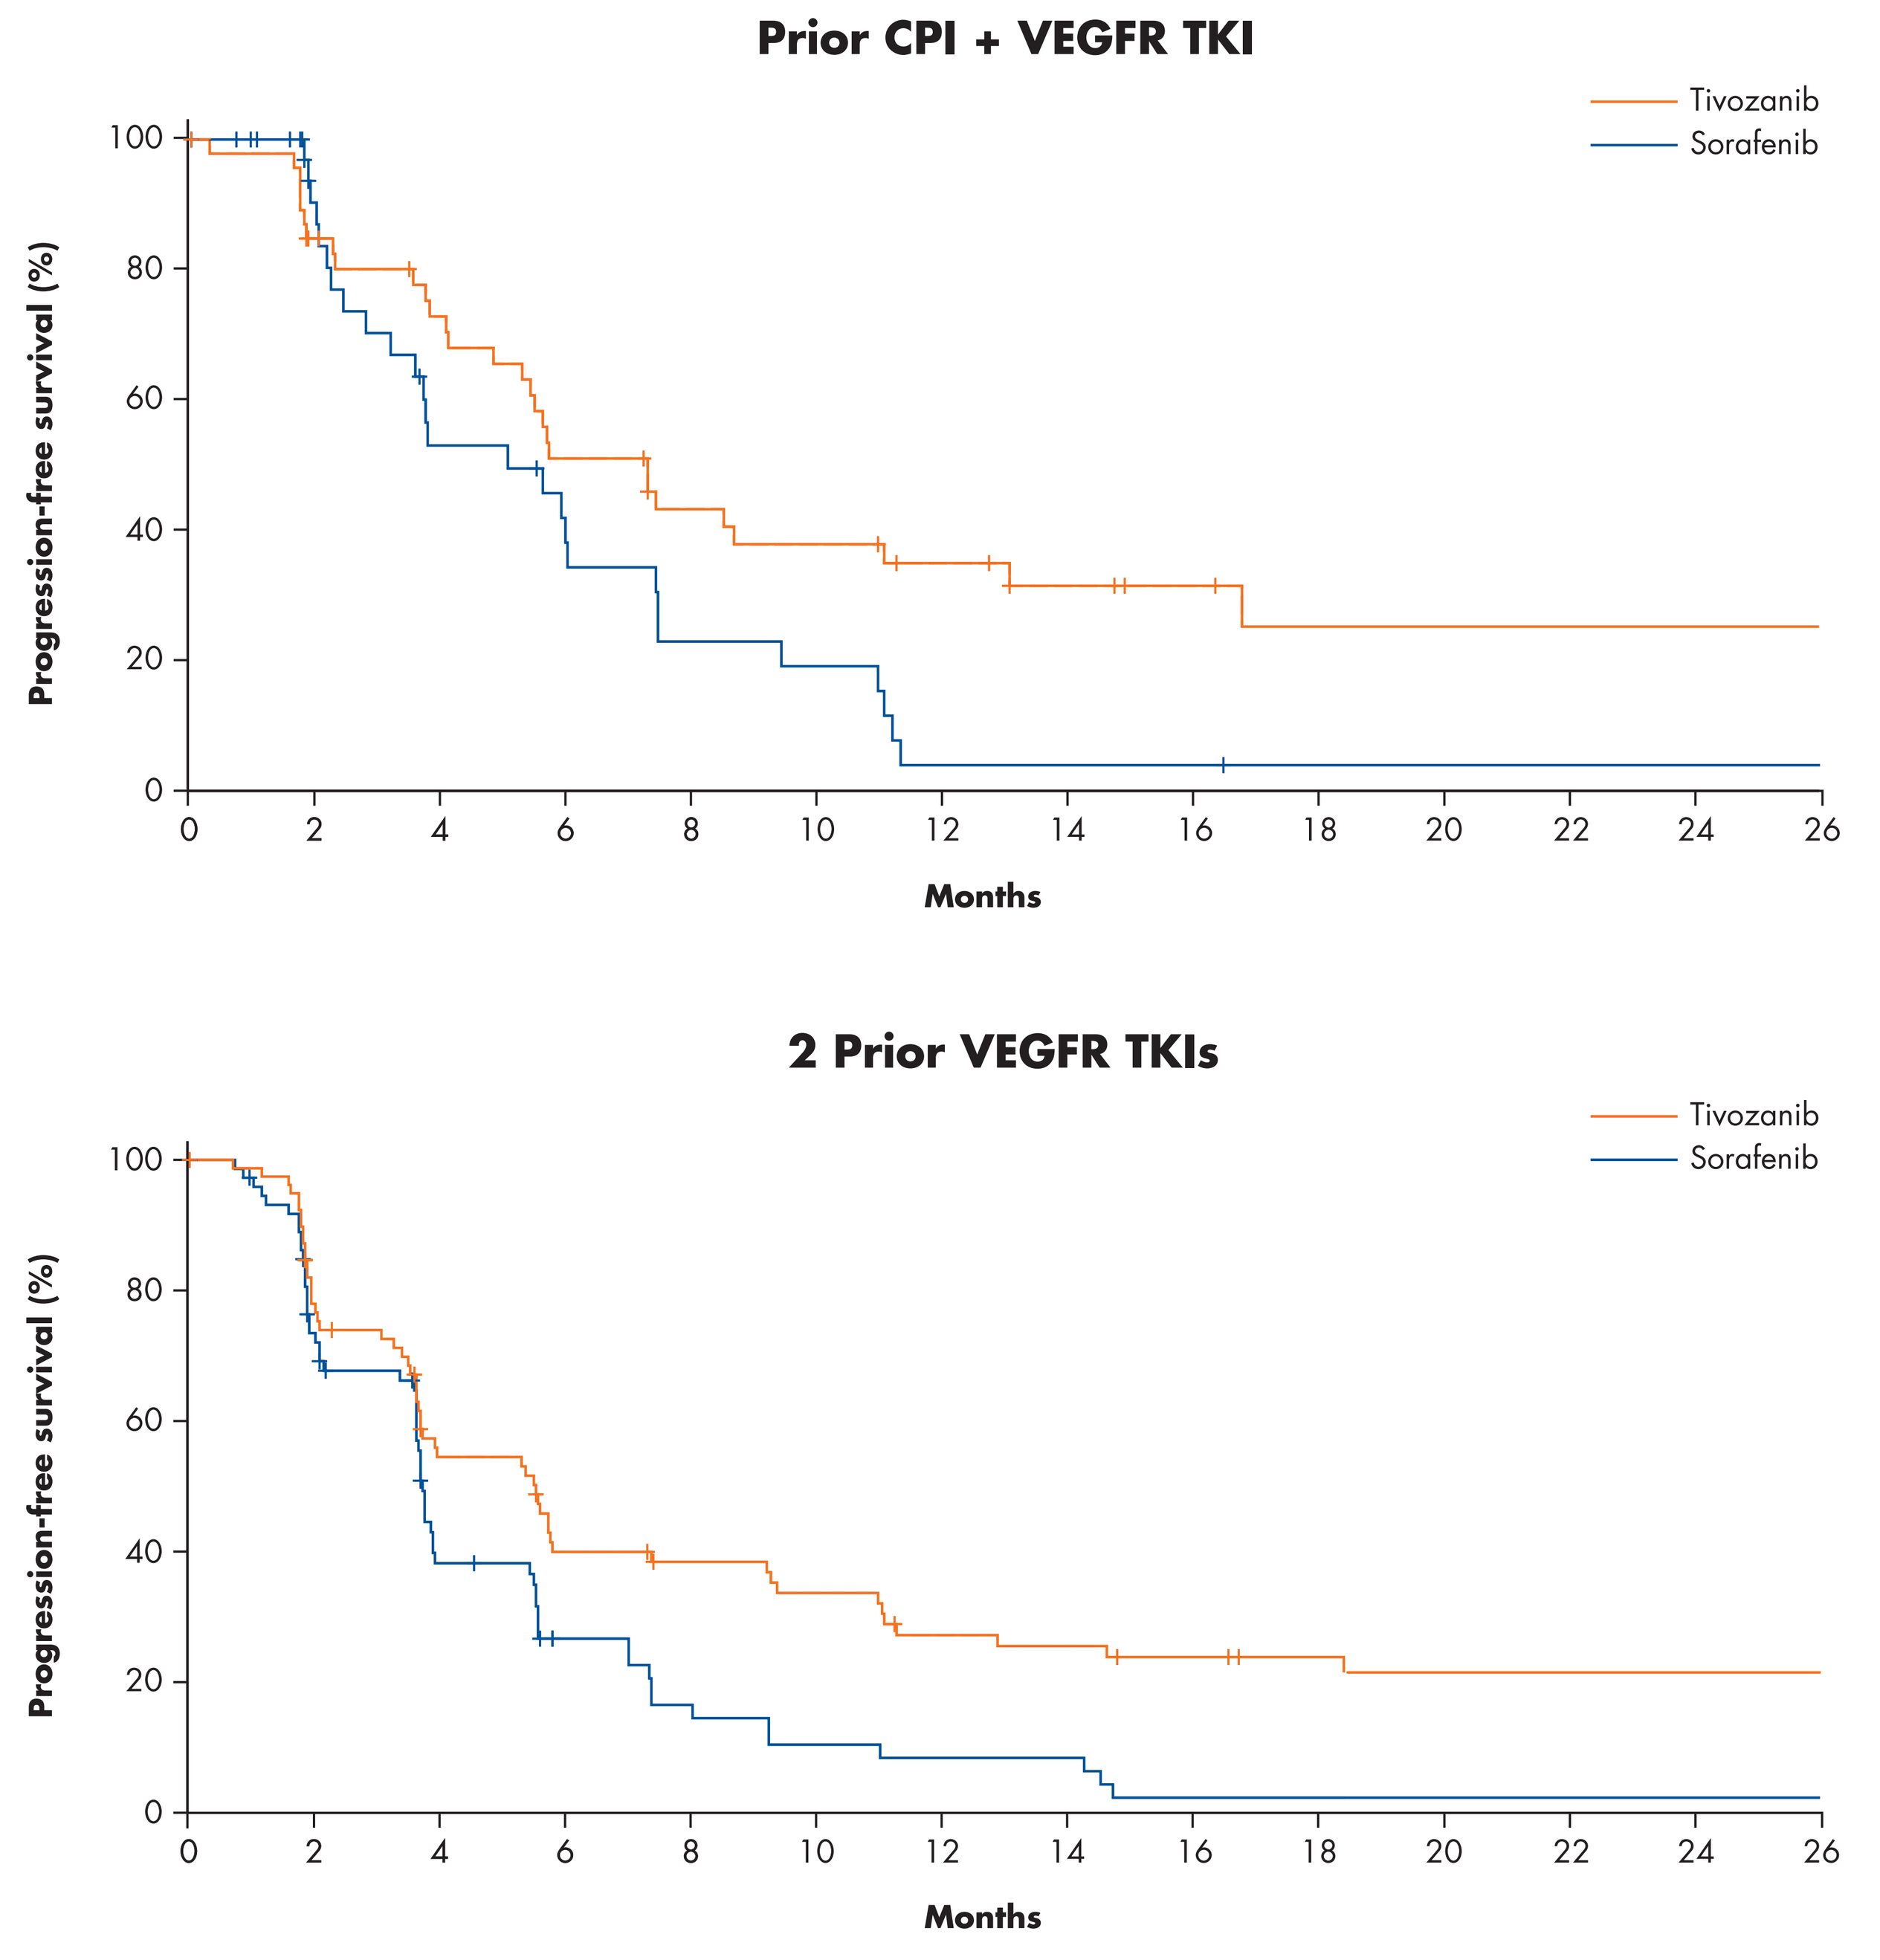

Prior CPI + VEGFR TKI
Tivozanib
Sorafenib
Progression-free survival (%)
100
80
60
40
20
0
0 2 4 6 8 10 12 14 16 18 20 22 24 26
Months
2 Prior VEGFR TKIs
Tivozanib
Sorafenib
Progression-free survival (%)
100
80
60
40
20
0
0 2 4 6 8 10 12 14 16 18 20 22 24 26
Months

PFS=progression-free survival.

Final OS – May 2020

- In the May 1, 2020, final OS analysis, with a median follow-up of 38 months for tivozanib and 40 months for sorafenib, 251 OS events occurred (125 for tivozanib and 126 for sorafenib)
 - No significant difference in OS was observed for tivozanib compared with sorafenib with an HR of 0.966 (95% CI: 0.75, 1.24; *P*=0.78) and median OS of 16.4 months vs. 19.2 months (**Figure 4**)
- At this time, final OS confirmation is pending

Figure 4. OS in the ITT Population

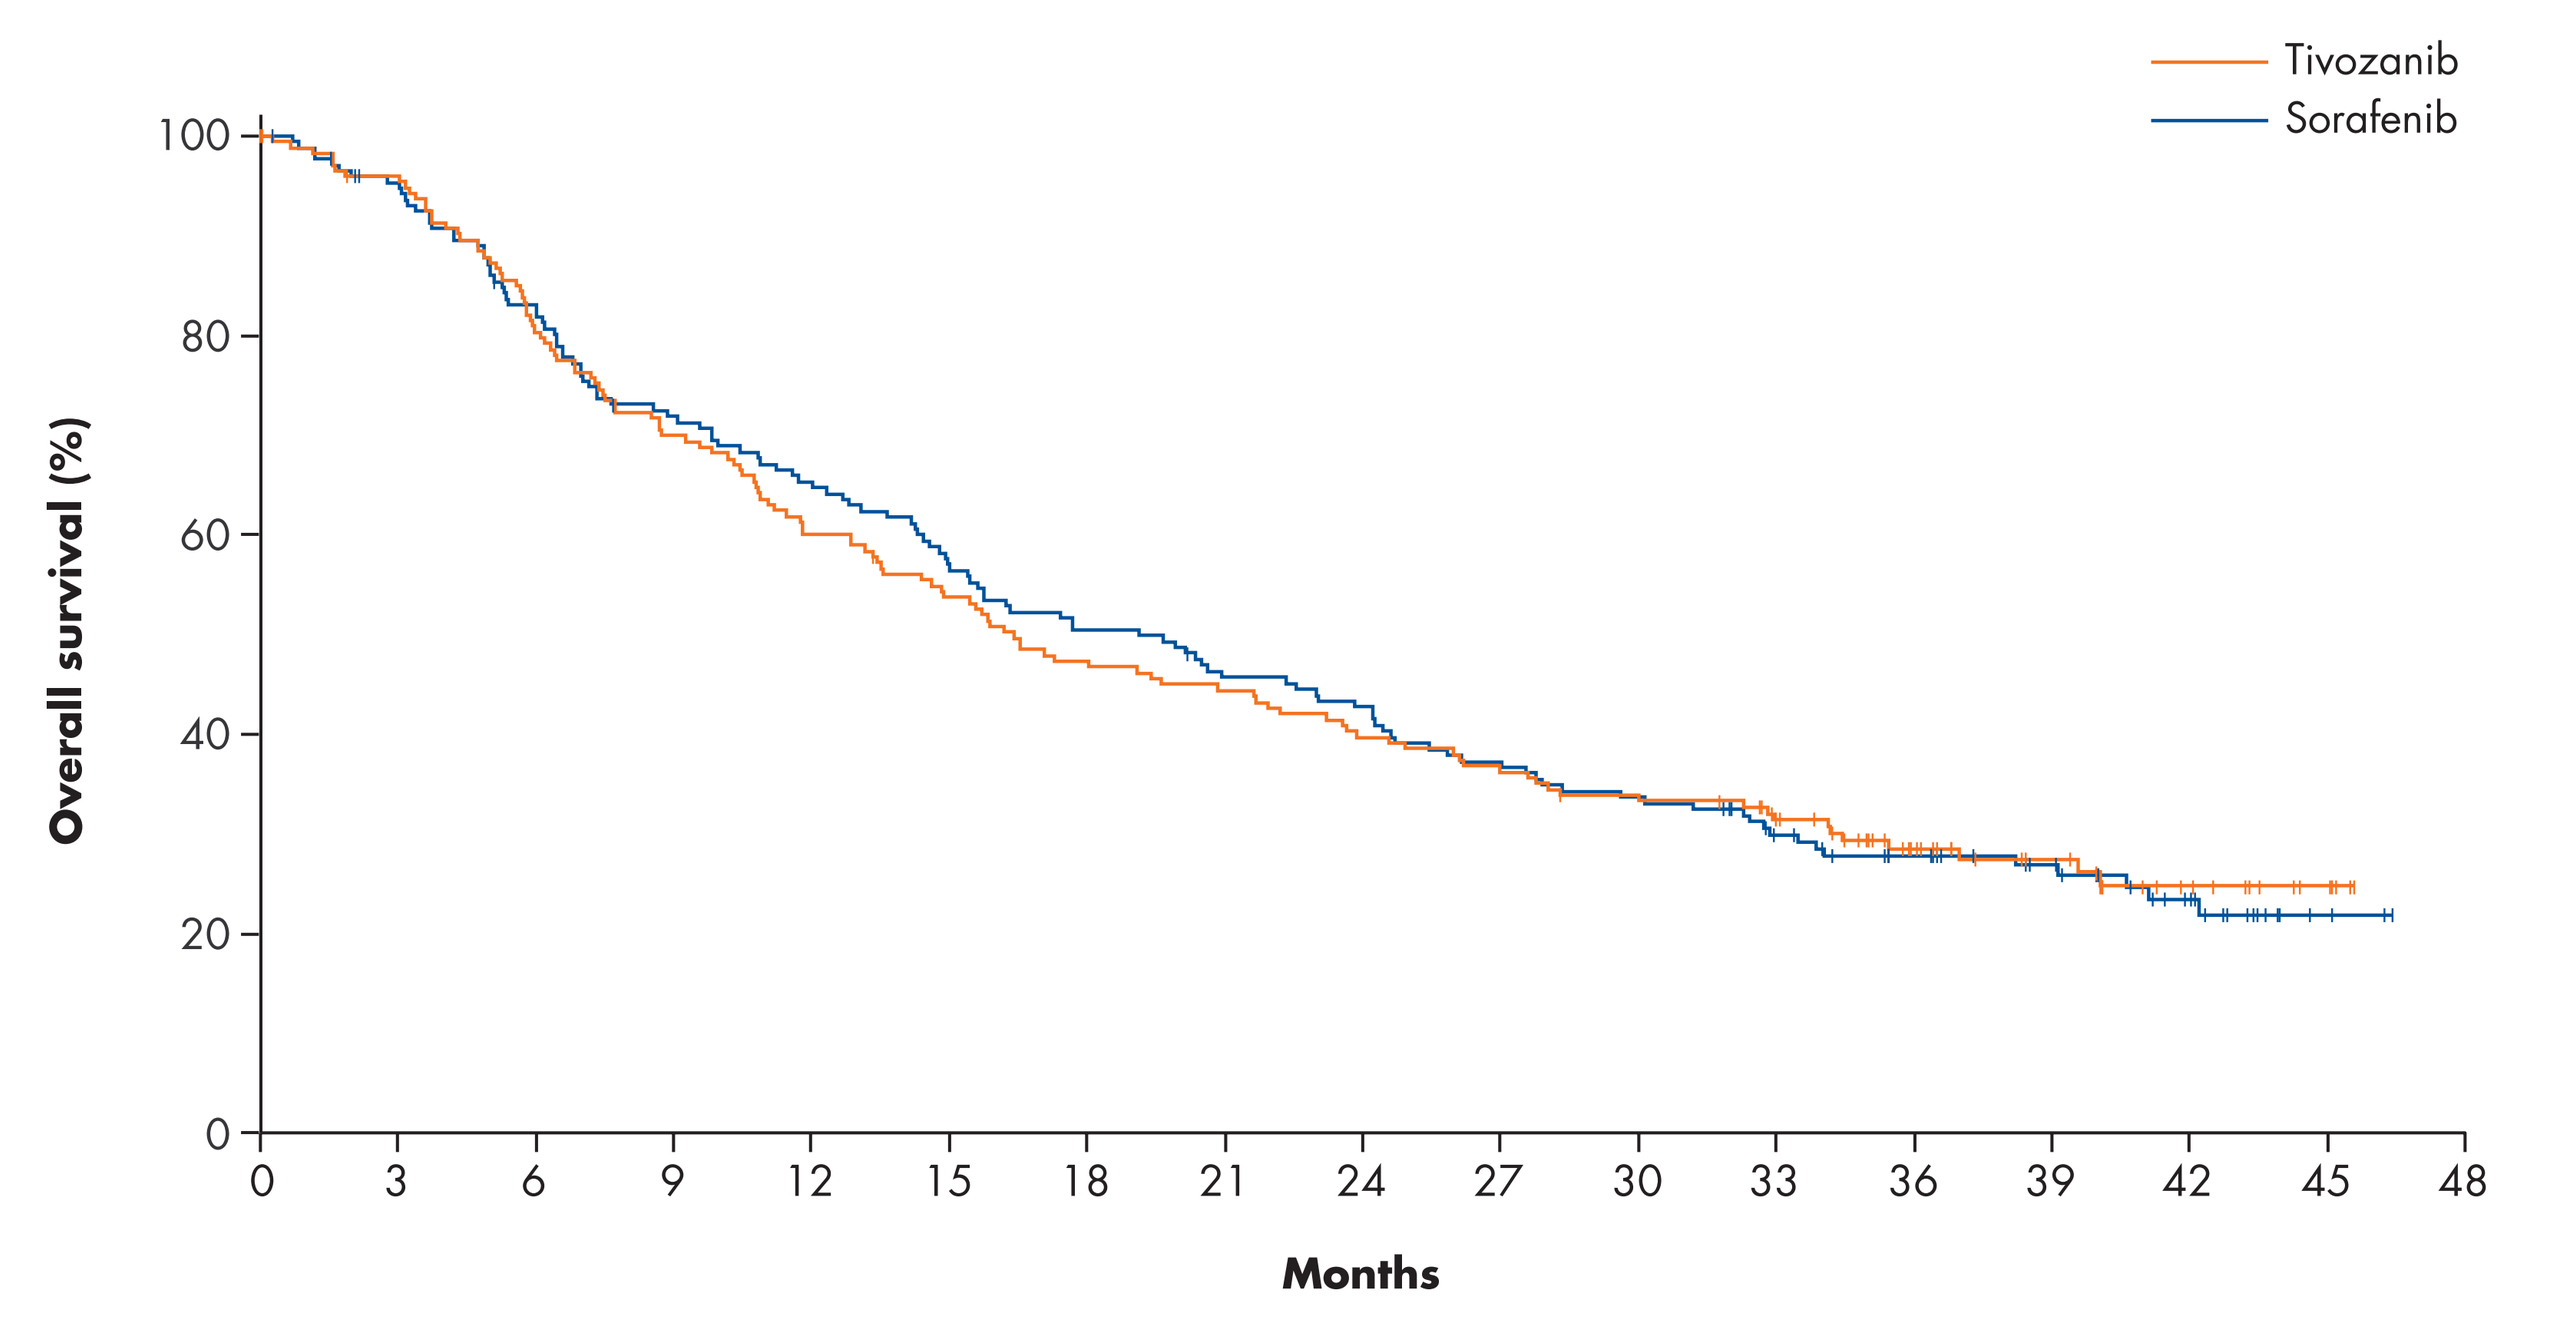

Tivozanib
Sorafenib
Overall survival (%)
100
80
60
40
20
0
0 3 6 9 12 15 18 21 24 27 30 33 36 39 42 45 48
Months

- OS in patients who received a prior CPI plus VEGFR TKI or 2 prior VEGFR TKIs is presented in **Table 2**

Table 2. OS in Select Patient Subgroups

	ITT (N=350)	Prior CPI + VEGFR TKI (n=91)	Two Prior VEGFR TKIs (n=159)
HR	0.97	0.84	0.99
95% CI	0.75-1.24	0.50-1.40	0.68-1.44

CI=confidence interval; CPI=checkpoint inhibitor; HR=hazard ratio; ITT=intent-to-treat; TKI=tyrosine kinase inhibitor; VEGFR=vascular endothelial growth factor receptor.

Response to Treatment

- Significantly more patients achieved a response in the tivozanib group than in the sorafenib group (18% vs. 8%; *P*=0.017; **Table 3**)
 - Median duration of response was not reached (NR; 95% CI: 12.9, NR) in the tivozanib group and 5.7 months (95% CI, 5.6, NR) in the sorafenib group

Table 3. Best Response to Treatment in Evaluable Patients[8]

	Tivozanib (n=175)	Sorafenib (n=175)
Best overall response		
CR	0	0
PR	31 (18)	14 (8)
SD	94 (54)	99 (57)
PD	37 (22)	32 (18)
NE	10 (6)	30 (17)
Objective response rate	31 (18)	14 (8)
95% CI	12.36, 24.19	4.44, 13.06
Disease control rate (CR+PR+SD)	125 (73)	113 (65)

Data are n (%) or median (95% CI).
CI=confidence interval; CR=complete response; NE=not evaluable; PD=progressive disease; PR=partial response; SD=stable disease.

Safety

- As of August 2019, treatment-related adverse events (AEs) were reported in 146 (84%) patients receiving tivozanib and 160 (94%) patients receiving sorafenib (**Table 4**)
 - The most common grade 3/4 treatment-related AE was hypertension, reported in 35 (20%) patients treated with tivozanib and 23 (14%) patients treated with sorafenib
 - Serious treatment-related AEs occurred in 19 (11%) patients receiving tivozanib and 17 (10%) patients receiving sorafenib
- Significantly fewer dose interruptions due to AEs were observed in patients treated with tivozanib (83 [48%]) than in patients treated with sorafenib (107 [63%]; *P*=0.0164)
- Significantly fewer dose reductions due to AEs were observed in patients treated with tivozanib (41 [24%]) than in patients receiving sorafenib (65 [38%]; *P*=0.0147)
- AEs led to discontinuation in 13 (8%) tivozanib-treated patients and 25 (15%) sorafenib-treated patients

Table 4. Treatment-Related AEs Reported in ≥20% of Patients[8]

	Tivozanib (n=173)		Sorafenib (n=170)	
Mean number of cycles initiated	12		7	
	All grades (%)	Grade 3/4 (%)	All grades (%)	Grade 3/4 (%)
Treatment-related AEs	84	46	94	55
Hypertension	38	21	25	14
Diarrhea	33	2	50	9
Fatigue	29	4	19	5
Decreased appetite	27	4	21	2
Dysphonia	24	1	8	0
Asthenia	22	5	17	4
Palmar-plantar erythrodysesthesia syndrome	16	1	39	10
Rash	5	0	24	8

Data are n (%).
AE=adverse event.

Conclusions

- In this final analysis of survival in the TIVO-3 trial, tivozanib demonstrated clinically meaningful and statistically significant improvement in PFS with similar OS in patients with highly relapsed or refractory metastatic RCC
 - Patients treated with a prior CPI and VEGFR TKI or 2 VEGFR TKIs derived the most PFS benefit from tivozanib (HR of 0.55 and 0.58, respectively) relative to sorafenib
- To our knowledge, TIVO-3 is the first randomized phase 3 study to show PFS superiority over another VEGFR TKI in the third- and fourth-line treatment setting, and is the first study to prospectively and comparatively evaluate treatment following CPI
- Consistent with other large studies in RCC comparing VEGFR TKIs, the OS HRs did not show significant OS differences between agents
- Collectively, these data support tivozanib as an evidence-based treatment option for patients with RCC, including for patients whose disease has progressed after previous CPI treatment

References

1. Escudier B et al. *N Engl J Med.* 2007;356:125-134.
2. Motzer RJ et al. *N Engl J Med.* 2007;356:115-124.
3. Rini B et al. *Clin Genitourin Cancer.* 2005;4:175-180.
4. Sternberg CN et al. *J Clin Oncol.* 2010;28:1061-1068.
5. Motzer RJ et al. *Lancet Oncol.* 2013;14:552-562.
6. Nakamura K et al. *Nat Med.* 1999;5:1359.
7. Eskens FA et al. *Clin Cancer Res.* 2011;17:7156-7163.
8. Rini B et al. *Lancet Oncol.* 2020;21:95-104.

PRESENTED AT THE AMERICAN SOCIETY OF CLINICAL ONCOLOGY ANNUAL MEETING; MAY 29-31, 2020.

E-mail: SPal@coh.org